Poster Number: 846

Contact: Aisling_Caffrey@uri.edu

Trends in *Acinetobacter baumannii* antibiotic resistance rates

Aisling R. Caffrey[1-4], Emily O'Neill[1,3], Haley Appaneal [1-4], Vrishali Lopes[1], Kerry L. LaPlante[1-3,5]

[1] Infectious Diseases Research Program, Providence Veterans Affairs Medical Center, Providence, RI, United States, [2]Center of Innovation in Long-Term Support Services, Providence Veterans Affairs Medical Center, Providence, RI, United States, [3]College of Pharmacy, University of Rhode Island, Kingston, RI, United States, [4] School of Public Health, Brown University, Providence, RI, [5]Warren Alpert Medical School of Brown University, Division of Infectious Diseases, Providence, RI

ABSTRACT

Background: Carbapenem-resistant *Acinetobacter baumannii* is described as an urgent threat by the Centers for Disease Control and Prevention and several older studies have indicated increasing resistance in *Acinetobacter.* We sought to describe these trends in the national Veterans Affairs (VA) Healthcare System.

Methods: We assessed *A. baumannii* positive clinical cultures collected from VA patients (≥ 18 years) from 2010 to 2018. We categorized cultures based on location at the time of collection: VA medical center (VAMC), community living center (CLC), or outpatient (Outpt). Multidrug resistance (MDR) and extensive drug resistance (XDR) were defined as resistance to > 1 drug in > 3 or all of the following categories, respectively: extended-spectrum cephalosporins (es-CS), fluoroquinolones (FQ), aminoglycosides (AMG), carbapenems (CARB), piperacillin/tazobactam (PIP/TAZ), and ampicillin/sulbactam (AMP/SUL). Joinpoint Software was used for regression analyses of trends over time and to estimate annual average percent changes (AAPC) with 95% confidence intervals.

Results: We identified 19,376 *A. baumannii* positive cultures over the study period (53% VAMCs, 4% CLCs, 43% Outpts), which represented 0.5% of all positive cultures in the VA. In VAMCs, the number of *A. baumannii* cultures decreased significantly by 11.9% per year. Of all positive cultures in VAMCs, the proportion that were A. baumannii decreased significantly by 5.4% per year. Similar trends were observed in CLCs, while Outpt cultures remained stable. Over the 9-year study period, resistance decreased significantly, with MDR decreasing by 10.2% per year and XDR decreasing by 9.4%. Carbapenem resistance decreased significantly by 4.9% per year in VAMCs (2010, 39%; 2018 28%) and 11.3% in Outpts (2010, 12%; 2018, 6%). Similar annual significant decreases were observed with AMG (9.4%), es-CS (1.4%), and FQ (7.4%) in VAMCs; es-CS (2.7%) and FQ (5.6%) in CLCs; and AMG (9.5%) and FQ (8.2%) in Outpts.

Conclusion: In the national VA Healthcare System, the prevalence of *A. baumannii* is decreasing, as is the resistance previously observed with this organism. MDR *A. baumannii* still made up one-third of cultures in VAMCs and CLCs in 2018, and thus remains a treatment challenge.

INTRODUCTION

Carbapenem-resistant *Acinetobacter baumannii* (CRAB) was upgraded to an urgent threat by the CDC in 2019. Despite reductions in CRAB infections in the United States, co-resistance to other antibiotics in CRAB is a major concern as standard-of-care antibiotics may be ineffective, presenting unique treatment challenges.

OBJECTIVE

Identify trends in resistance rates of *A. baumannii* isolates across the Veterans Affairs (VA) Healthcare System nationally.

METHODS

- *A. baumannii* clinical cultures, 2010 to 2018 collected in VA medical centers (VAMC), community living centers (CLC), or outpatient clinics (Outpt).
- For resistance, included the first isolate per patient, per facility, per year. Antibiotic susceptibility based on latest Clinical Laboratory Standards Institute breakpoints to numeric minimum inhibitory concentrations (MIC) data were available. Where MIC values were not available, we used the reported textual interpretations (i.e., resistant [R], intermediate [I], or susceptible [S]).
- Time trends assessed with Joinpoint (JP) regression, to calculate average annual percent changes (AAPC) and 95% confidence intervals (CI).

RESULTS

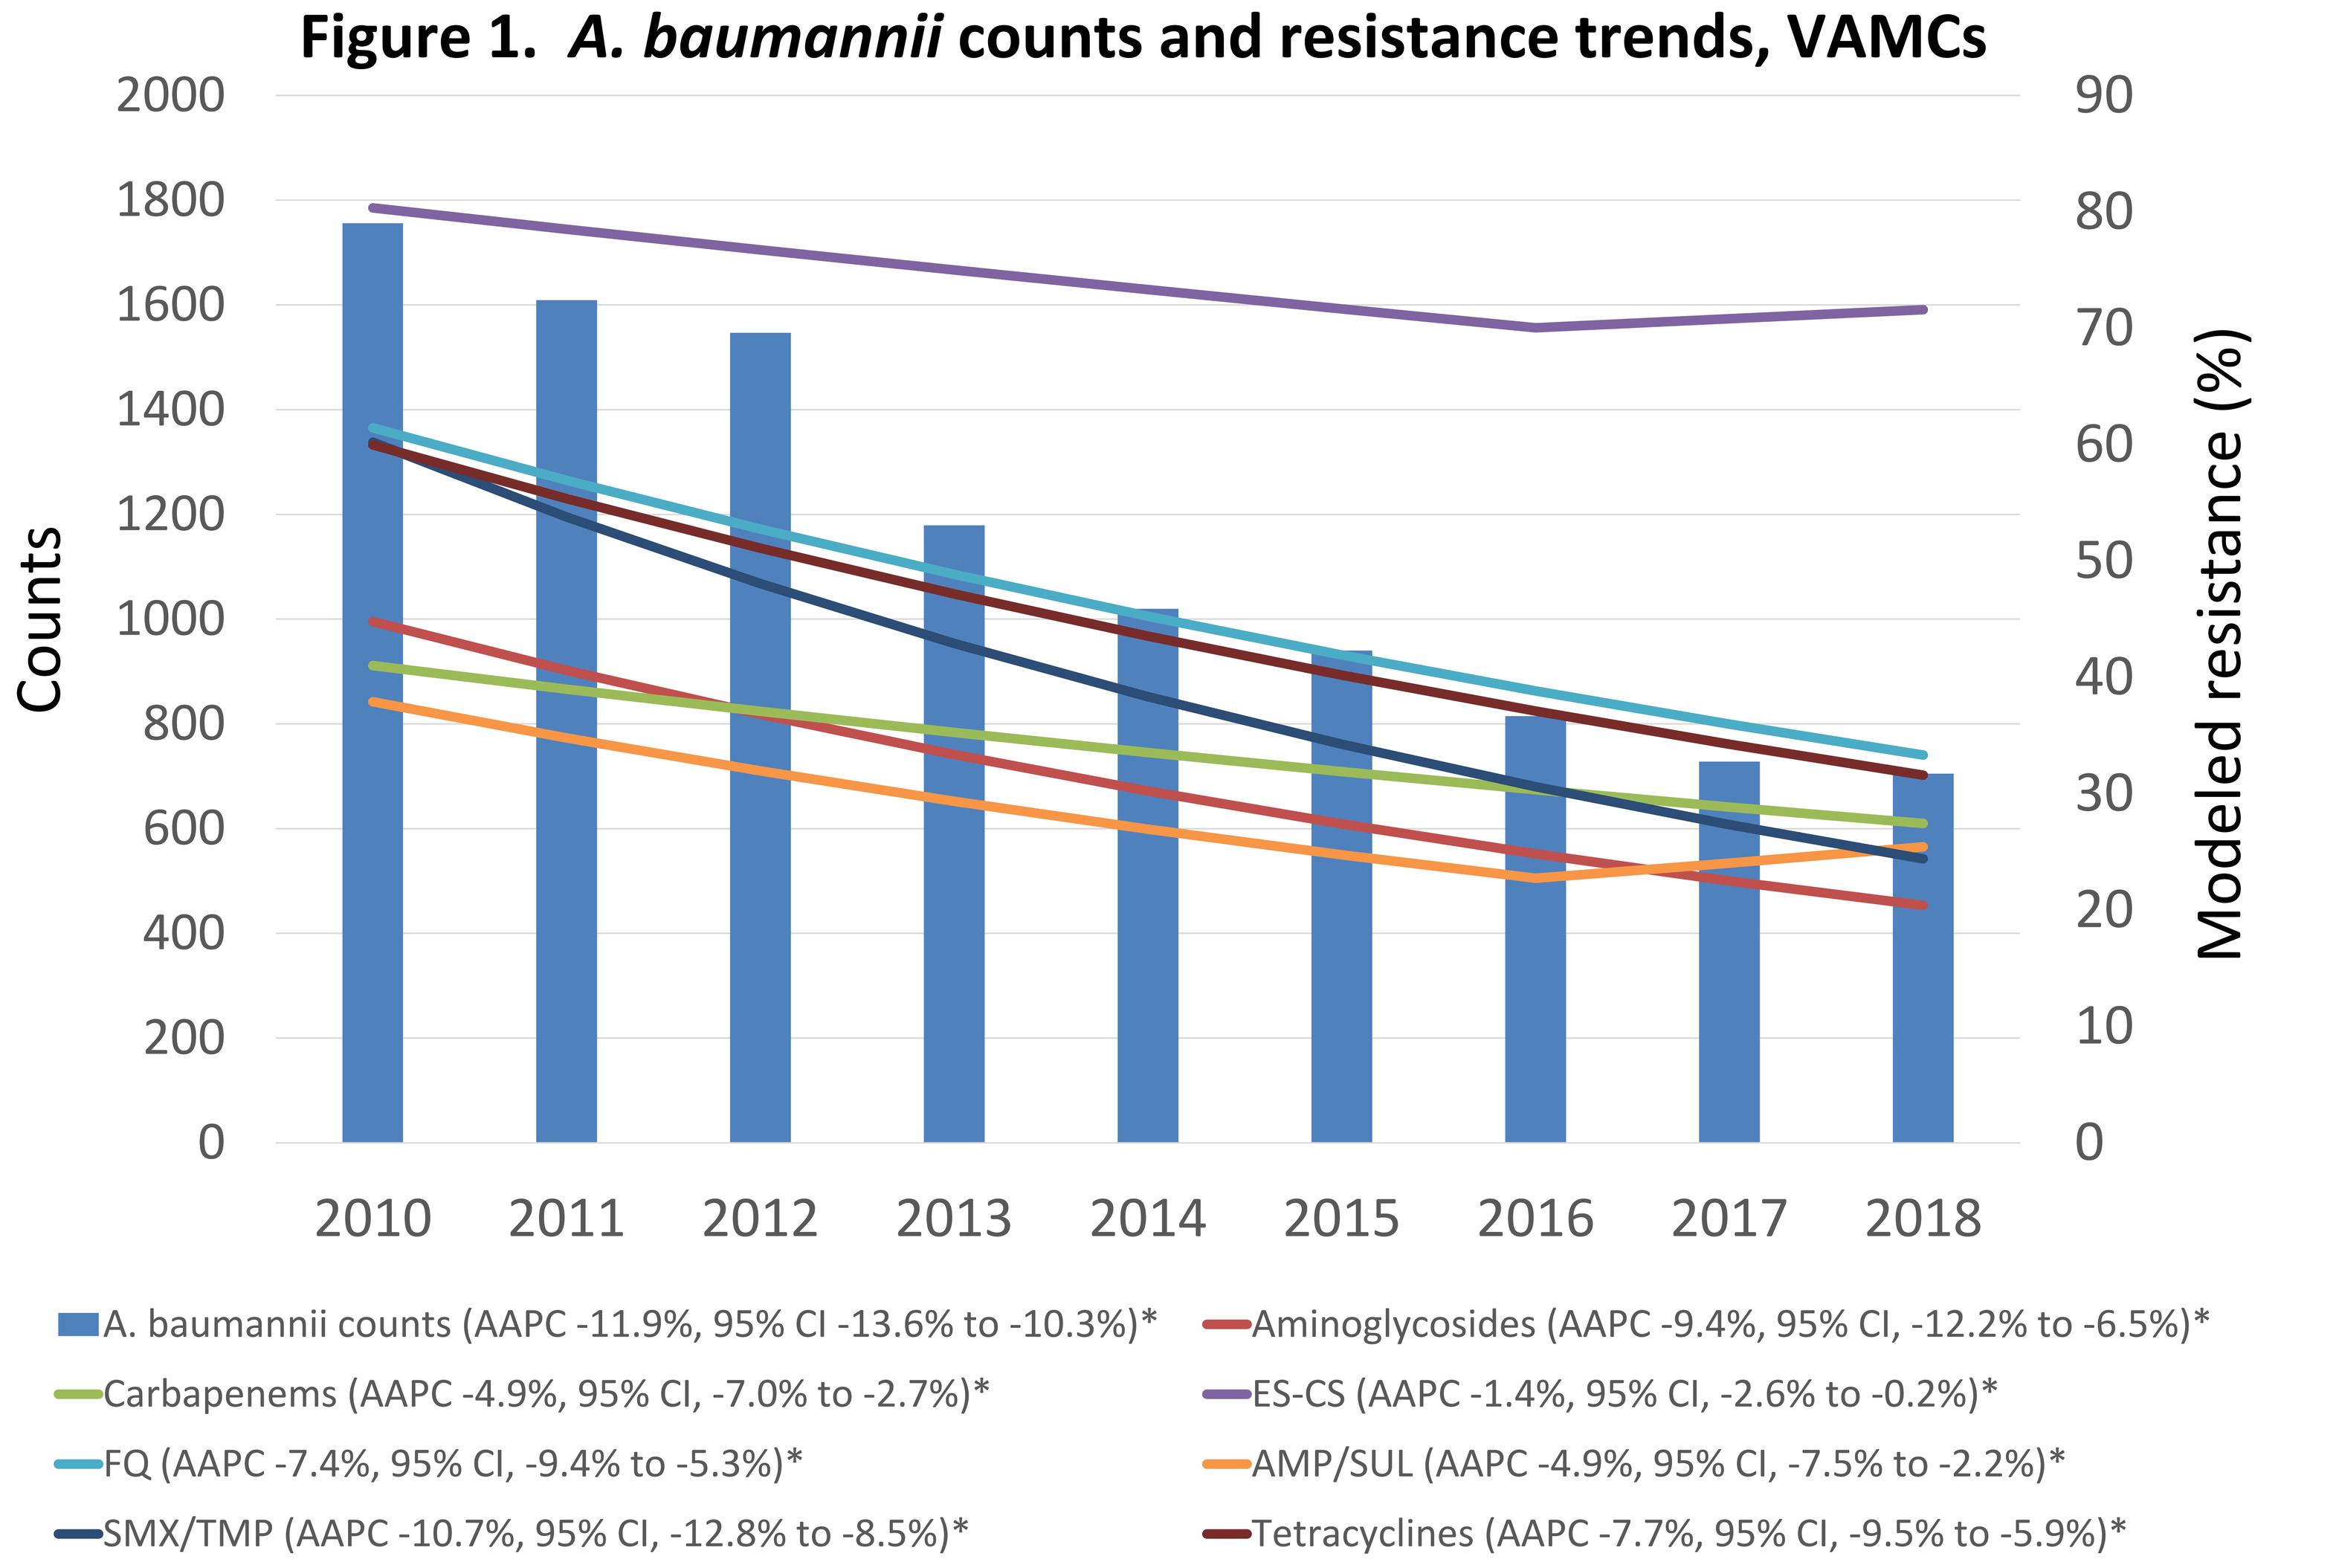
Counts
Modeled resistance (%)
2010
2011
2012
2013
2014
2015
2016
2017
2018
A. baumannii counts (AAPC -11.9%, 95% CI -13.6% to -10.3%)*
Aminoglycosides (AAPC -9.4%, 95% CI, -12.2% to -6.5%)*
Carbapenems (AAPC -4.9%, 95% CI, -7.0% to -2.7%)*
ES-CS (AAPC -1.4%, 95% CI, -2.6% to -0.2%)*
FQ (AAPC -7.4%, 95% CI, -9.4% to -5.3%)*
AMP/SUL (AAPC -4.9%, 95% CI, -7.5% to -2.2%)*
SMX/TMP (AAPC -10.7%, 95% CI, -12.8% to -8.5%)*
Tetracyclines (AAPC -7.7%, 95% CI, -9.5% to -5.9%)*

Figure 1. ***A. baumannii*** **counts and resistance trends, VAMCs**

* Indicates p-value <0.05. Aminoglycosides (amikacin, gentamicin, tobramycin), Carbapenems (imipenem, meropenem, doripenem), ES-CS = Extended-spectrum Cephalosporins (cefepime, ceftazidime, cefotaxime, ceftriaxone), Fluoroquinolones (ciprofloxacin, levofloxacin), Antipseudomonal penicillin's + β-lactamase inhibitors (piperacillin/tazobactam, clavulanate/ticarcillin), Amp/Sul = Ampicillin/Sulbactam, SMX/TMP = Sulfamethoxazole/Trimethoprim, Tetracyclines (tetracycline, minocycline, doxyclycline).

RESULTS

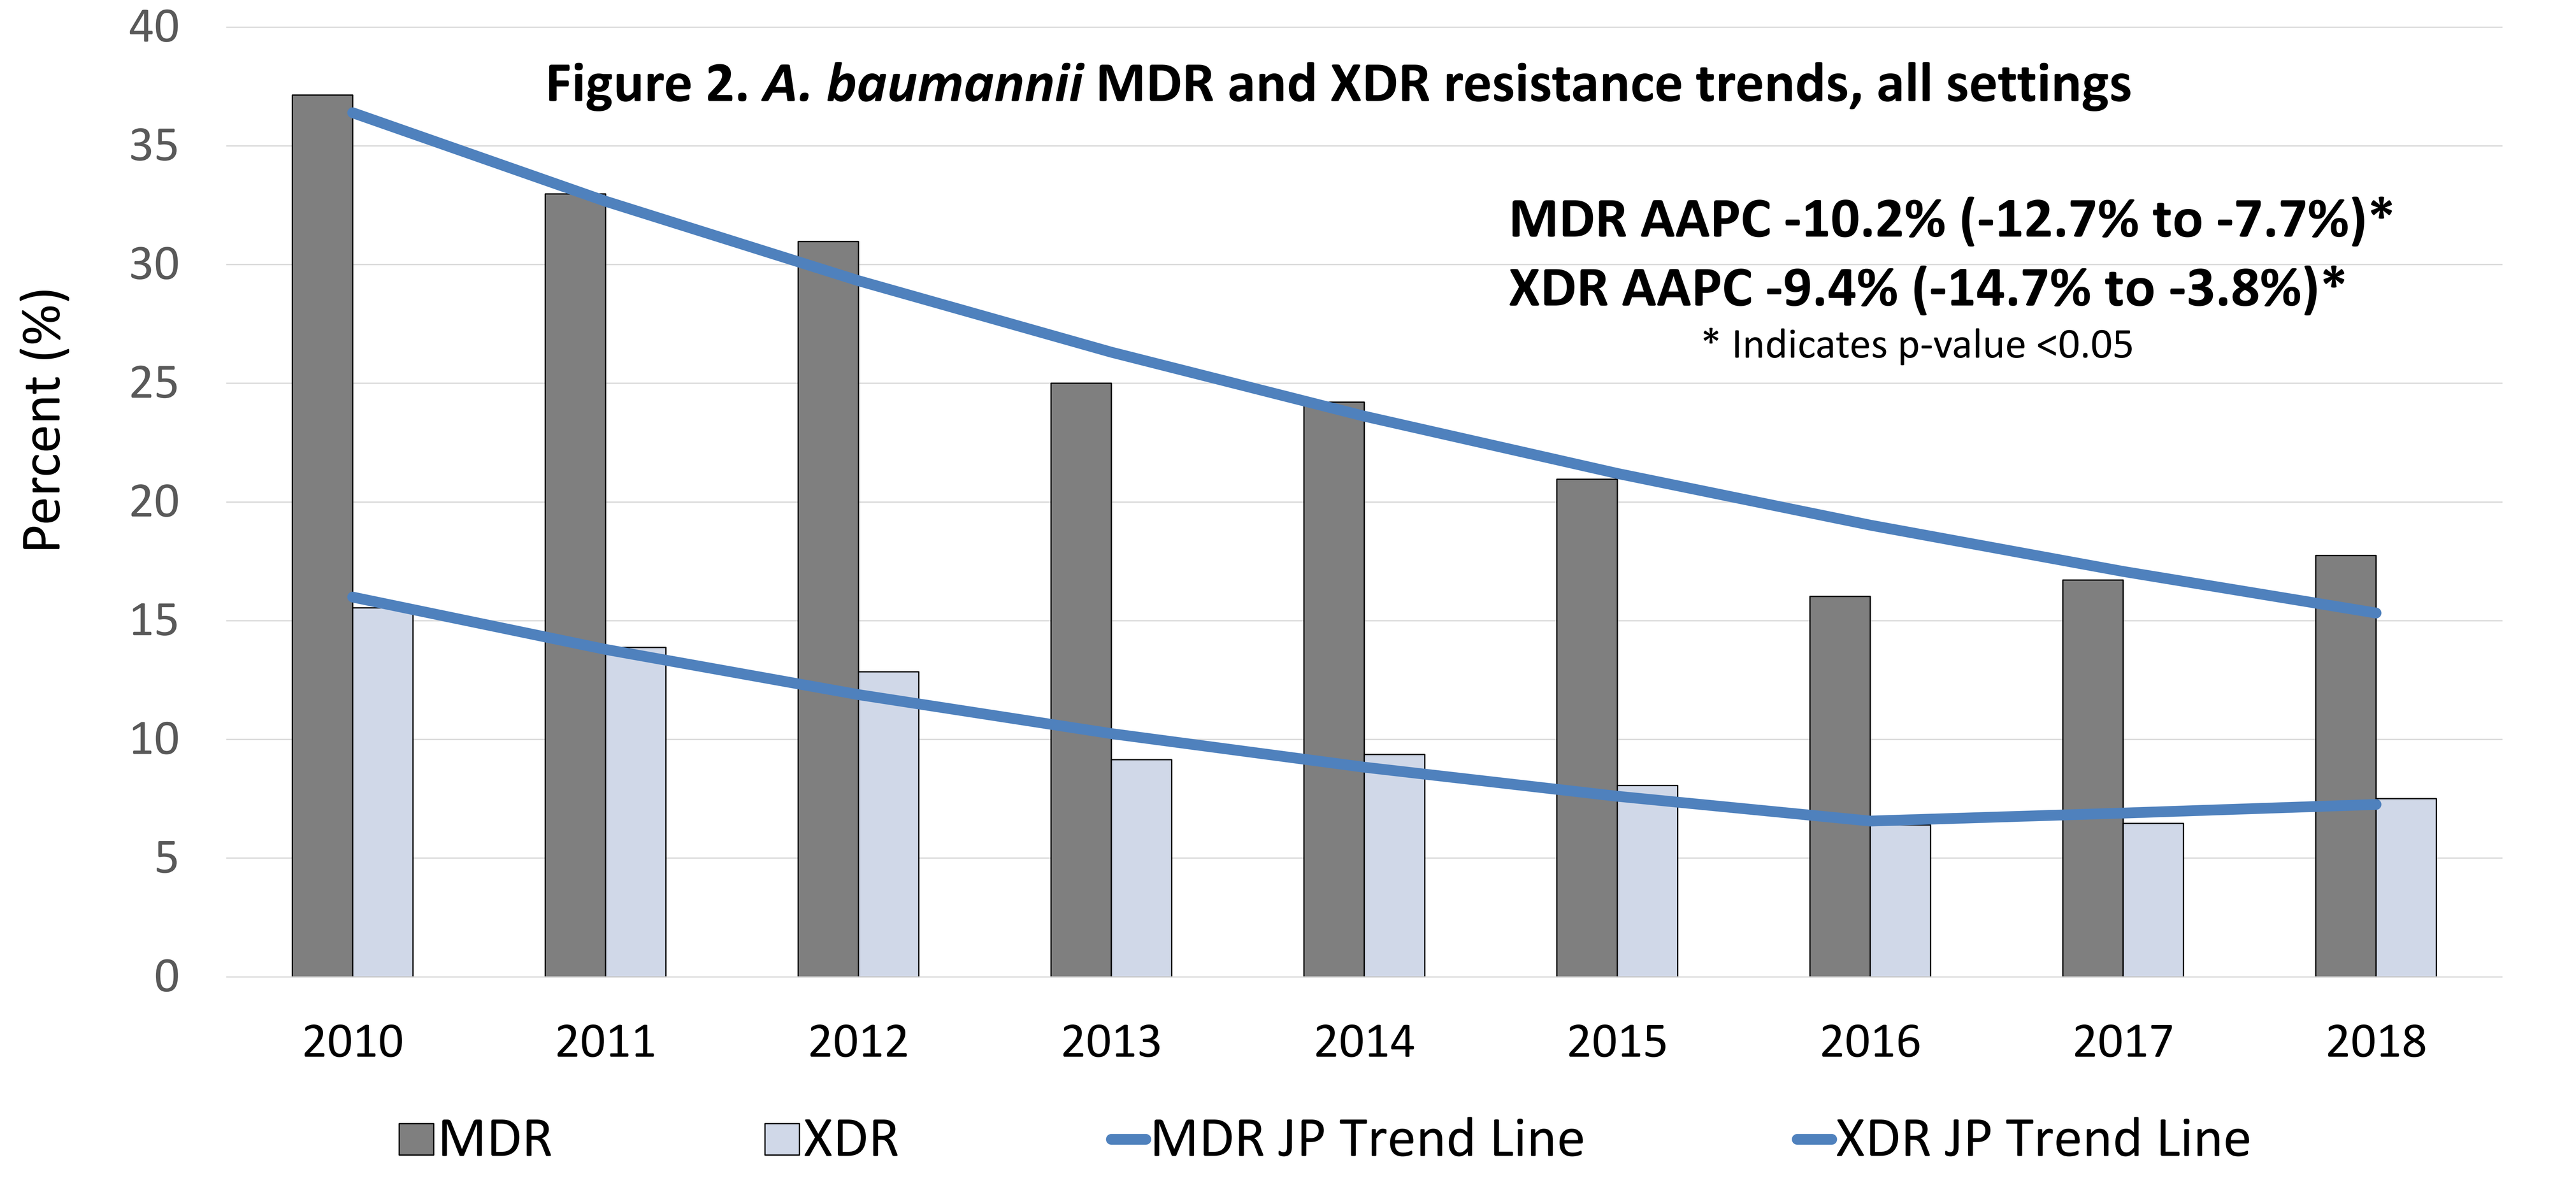
Percent (%)
MDR AAPC -10.2% (-12.7% to -7.7%)*
XDR AAPC -9.4% (-14.7% to -3.8%)*
* Indicates p-value <0.05
2010
2011
2012
2013
2014
2015
2016
2017
2018
MDR
XDR
MDR JP Trend Line
XDR JP Trend Line

Figure 2. ***A. baumannii*** **MDR and XDR resistance trends, all settings**

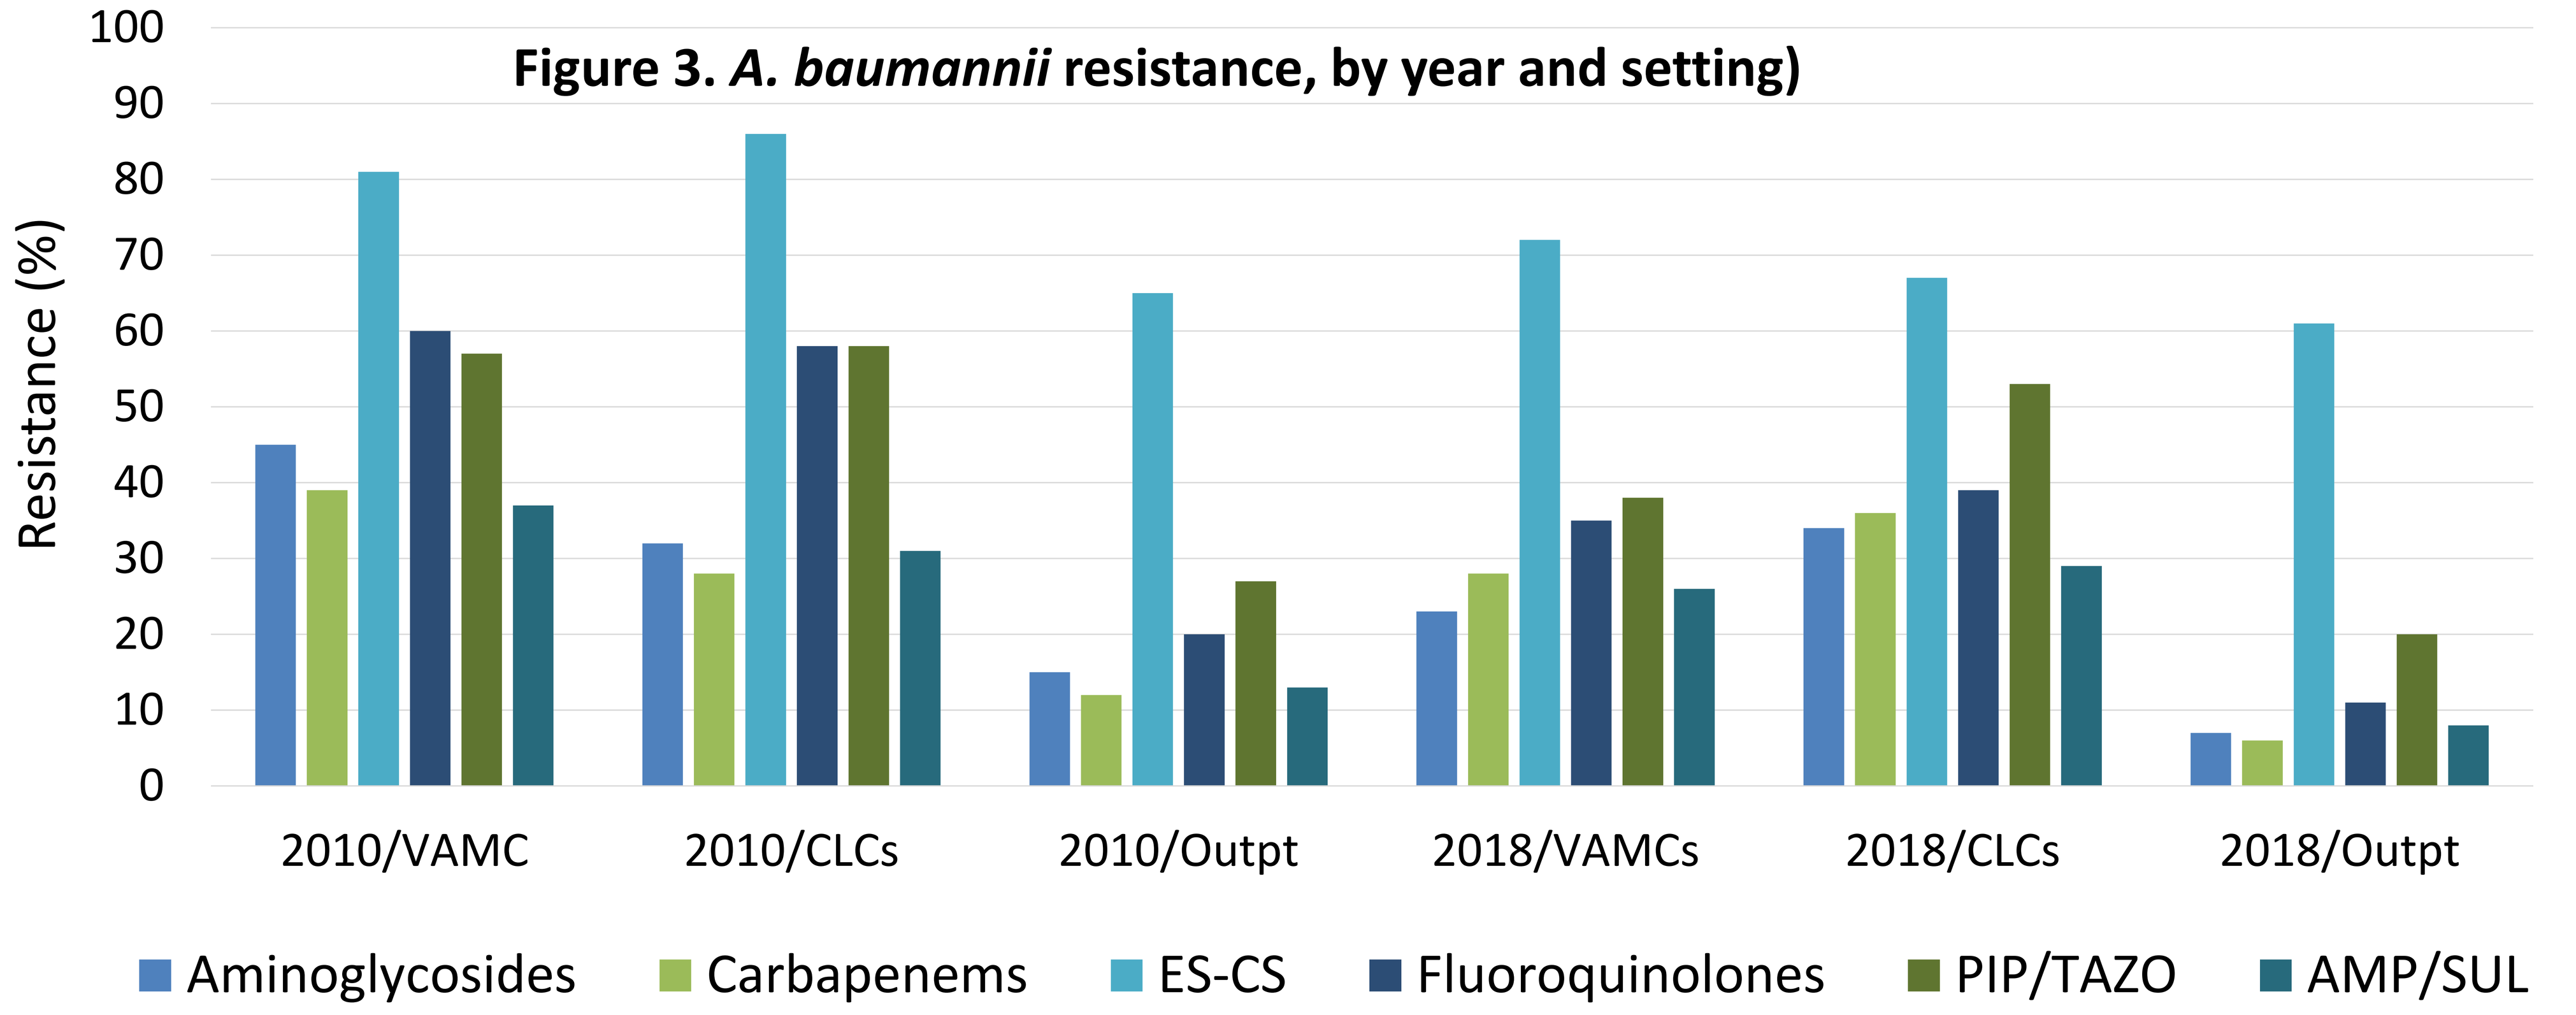
Resistance (%)
2010/VAMC
2010/CLCs
2010/Outpt
2018/VAMCs
2018/CLCs
2018/Outpt
Aminoglycosides
Carbapenems
ES-CS
Fluoroquinolones
PIP/TAZO
AMP/SUL

Figure 3. ***A. baumannii*** **resistance, by year and setting)**

Of isolates tested for susceptibility, ES-CS = Extended-spectrum Cephalosporins, PIP/TAZO = piperacillin/tazobactam, Amp/Sul = Ampicillin/Sulbactam

CONCLUSIONS

We observed significant decreases in *A. baumannii* clinical cultures among VA inpatient populations over our recent 9 year study period. Significant decreases were observed in *A. baumannii* resistance, including MDR (10.2%), XDR (9.4%), CRAB (4.9% VAMC, 11.3% Outpt). In 2018, CRAB was 28% in VAMCs and 36% in long-term care, which present challenges to effective treatment despite these improvements in resistance.

Citation: CDC. Antibiotic Resistance Threats in the United States, 2019. Atlanta, GA: U.S. Department of Health and Human Services, CDC; 2019.
Acknowledgements: The information presented are those of the authors and do not necessarily reflect the position or policy of the United States Department of Veterans Affairs. **Funding:** This work was funded in part by Shinogi, Inc. **Conflicts of Interest:** KLL has received research funding or is an advisor/consultant for Merck, Pfizer Pharmaceuticals, Ocean Spray Cranberries, Inc., Nabriva Therapeutics US, Inc., Melinta Therapeutics, Inc., and Tetraphase Pharmaceuticals. ARC has received research funding from Pfizer, Merck (Cubist), and Shionogi. No other financial disclosures.

ID Week 2020, October 21st -25th 2020, Virtual Event